EMERGE Study: An Open-Label Evaluation of the Efficacy and Safety of AXS-07 (MoSEIC™ Meloxicam–Rizatriptan) for Migraine in Adults With Prior Inadequate Response to an Oral CGRP Inhibitor

Peter McAllister,[1] Stewart Tepper,[2] Angad Chhabra,[3] Caroline Streicher,[3] Gregory Parks,[3] Herriot Tabuteau[3]

[1]New England Institute for Neurology and Headache, Stamford, CT, USA; [2]Geisel School of Medicine at Dartmouth, New Hanover, NH, USA; [3]Axsome Therapeutics, New York, NY, USA

Key Questions

What is AXS-07?
What is the EMERGE study?

Conclusions

EMERGE is the first study to investigate the efficacy, tolerability, and safety of AXS-07 for acute treatment of migraine in adults with a history of migraine attacks and inadequate response to oral CGRP inhibitors used for acute treatment.

Acknowledgments

This study was sponsored by Axsome Therapeutics. Medical writing support was provided by Iain McDonald, PhD, CMPP and Lori Kornberg, PhD, of Envision Pharma Group, and was funded by Axsome Therapeutics.

Disclosures

PM, ST are consultants for Axsome Therapeutics.

AC, CS, GP, and HT are employees of Axsome Therapeutics and may hold stock or stock options.

Please scan this QR code with your smartphone app to view an electronic version of this poster. If you do not have a smartphone, access the poster via the internet at: axsomecongresshub.com/aan2024

Copies of this poster obtained through QR code are for personal use only and may not be reproduced without permission of the authors.

American Academy of Neurology (AAN) 2024 Annual Meeting, Denver, CO, USA
April 13–18, 2024

Introduction

- Surveys of patients with migraine have identified dissatisfaction with their current treatment,[1,2] with even those who are satisfied expressing a willingness to try a new migraine treatment.[3]
- Calcitonin gene-related peptide (CGRP) inhibitors have been recently introduced for the treatment of acute attacks; however, a significant percentage of patients treated with these agents are not free from pain or their most bothersome symptom (MBS) 2-hours post dose.[4]
- There remains an unmet need for effective agents for the acute treatment of migraine attacks, particularly in patients who have failed treatment with CGRP inhibitors.

Overview of AXS-07

- AXS-07, a novel, oral, rapidly absorbed, investigational medicine, consists of 20 mg MoSEIC™ meloxicam and 10 mg rizatriptan.[5,6]
 – MoSEIC™ (Molecular Solubility Enhanced Inclusion Complex) delivery technology increases the solubility and absorption speed of oral meloxicam with maintenance of the extended plasma half-life.[6]
- AXS-07 exhibits multiple mechanisms of action (**Figure 1**):[5,7]
 – Inhibition of CGRP release and reversal of CGRP mediated vasodilation (rizatriptan).
 – Inhibition of neuroinflammation and central sensitization (MoSEIC™ meloxicam).
 – Inhibition of pain signal transmission (rizatriptan).
- Median time to reach therapeutic plasma concentration of MoSEIC™ meloxicam is 17 minutes.[6]

Figure 1. Mechanism of Action of AXS-07[5]

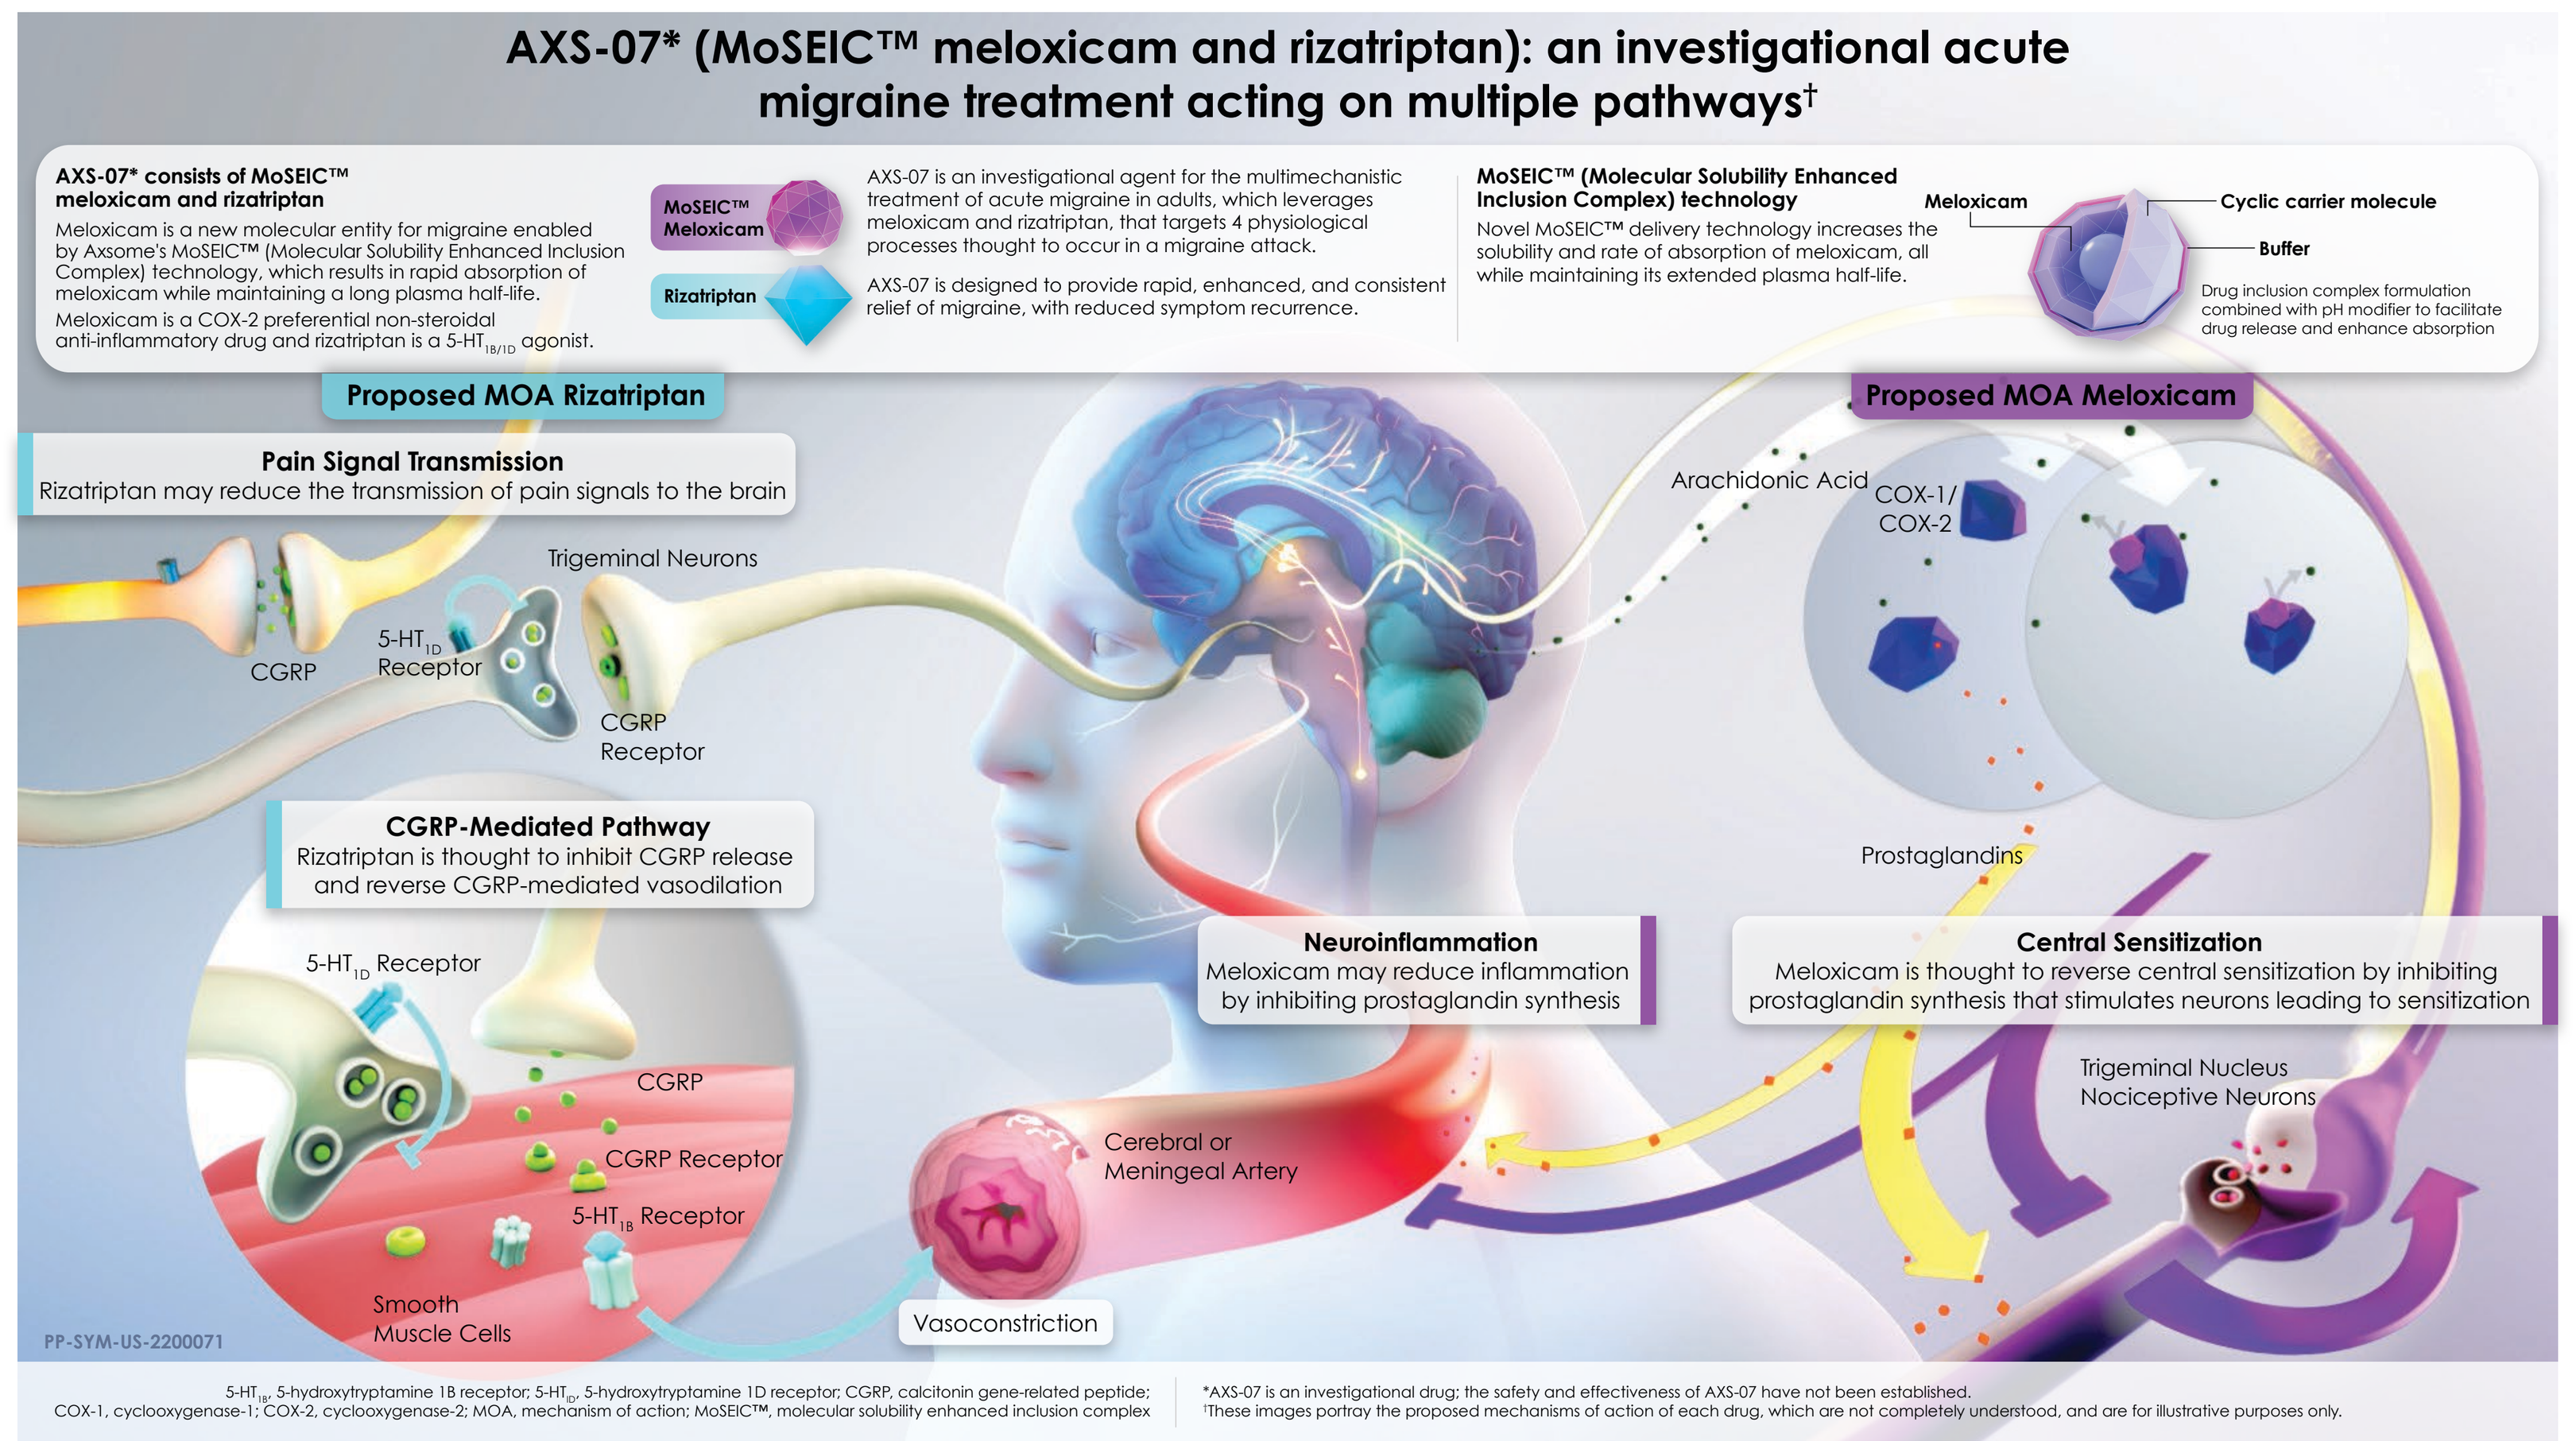
AXS-07* (MoSEIC™ meloxicam and rizatriptan): an investigational acute migraine treatment acting on multiple pathways†
AXS-07* consists of MoSEIC™ meloxicam and rizatriptan
MoSEIC™ Meloxicam
Rizatriptan
MoSEIC™ (Molecular Solubility Enhanced Inclusion Complex) technology
Meloxicam
Cyclic carrier molecule
Buffer
Proposed MOA Rizatriptan
Pain Signal Transmission
Rizatriptan may reduce the transmission of pain signals to the brain
Trigeminal Neurons
CGRP
CGRP Receptor
CGRP-Mediated Pathway
Rizatriptan is thought to inhibit CGRP release and reverse CGRP-mediated vasodilation
CGRP Receptor
Smooth Muscle Cells
Cerebral or Meningeal Artery
Vasoconstriction
Proposed MOA Meloxicam
Arachidonic Acid
COX-1/ COX-2
Prostaglandins
Neuroinflammation
Meloxicam may reduce inflammation by inhibiting prostaglandin synthesis
Central Sensitization
Meloxicam is thought to reverse central sensitization by inhibiting prostaglandin synthesis that stimulates neurons leading to sensitization
Trigeminal Nucleus Nociceptive Neurons

Source: Adapted from https://www.axsome.com/axs-portfolio/pipeline/about-axs-07

- In 2 phase 3 clinical trials, AXS-07 has shown statistically significant elimination of pain for patients with migraine relative to those on placebo and to active controls.[7-9]
 – MOMENTUM (NCT03896009) was a randomized, phase 3 double-blind,4-arm, parallel group, single-dose, placebo-controlled trial with a factorial study design in participants with a history of inadequate response to acute treatment of migraine, in which AXS-07 improved clinical outcomes compared with MoSEIC™ meloxicam, rizatriptan, and placebo (**Table 1**).[7,8]
 – In INTERCEPT (NCT04163185), a double-blind, single-dose, placebo-controlled phase 3 trial, patients were administered AXS-07 at the earliest sign of migraine pain, while the pain was mild. In INTERCEPT, AXS-07 improved clinical outcomes compared with placebo (**Table 1**).[9]
 – The safety findings across both trials indicated AXS-07 was well tolerated.[7-9]

Table 1. Co-primary Endpoints in Phase 3 Trials of AXS-07 (% Participants)

	AXS-07	Placebo	Difference	*P* value
MOMENTUM[7,8]	N=428	N=209		
Pain freedom at Hour 2	19.9%	6.7%	-13.2%	<0.001
Absence of MBS at Hour 2	36.9%	24.4%	-12.5%	0.002
INTERCEPT[9]	N=132	N=135		
Pain freedom at Hour 2	32.6%	16.3%	-16.3%	0.002
Absence of MBS at Hour 2	43.9%	26.7%	-17.3%	0.003

Sources: Jones A, et al. Poster presented at AAN 2022. P13-005; Jones, et al. Oral Presentation at AHS 2021; Jones, et al. Poster presented at AAN 2020
MBS, most bothersome symptom

Objective

- To describe the methodology of a phase 3 multicenter, open-label trial (EMERGE, NCT05550207) evaluating the efficacy and safety of AXS-07 in the acute treatment of migraine headache in adults with a prior inadequate response to an oral CGRP inhibitor.

Methods and Study Design

- EMERGE is a multicenter, phase 3, single-group trial (NCT05550207) evaluating the efficacy and safety of AXS-07 in the acute treatment of migraine headache in adults with a prior inadequate response to an oral CGRP inhibitor (**Tables 2-3**).

Table 2. Key Study Eligibility Criteria

Inclusion Criteria	Exclusion Criteria
Male or female, ≥18 years of age	Pregnant, breastfeeding, or planning to become pregnant during the study
Established diagnosis of migraine with or without aura	Previously received any investigational drug or device or investigational therapy within 30 days before screening
Inadequate response to oral CGRP inhibitors for acute treatment of migraine	

Source: https://www.clinicaltrials.gov/study/NCT05550207
CGRP, calcitonin gene-related peptide

Table 3. Primary Outcome Measures

Outcome	Measure	Time Point
% of participants with headache pain relief	Relief of headache pain without the use of rescue medication	Hour 2 after taking AXS-07
% of participants with absence of the MBS	Absence of the MBS, defined at migraine onset	Hour 2 after taking AXS-07

Source: https://www.clinicaltrials.gov/study/NCT05550207
MBS, most bothersome symptom

- AXS-07 tablets will be taken orally for up to 8 weeks for treatment of the participant's next 4 migraine attacks.
- ~ 100 participants were targeted for enrollment. Enrollment began in August 2022 and is ongoing across approximately 31 clinical sites in the United States (**Figure 2**).

Figure 2. Map of Study Sites

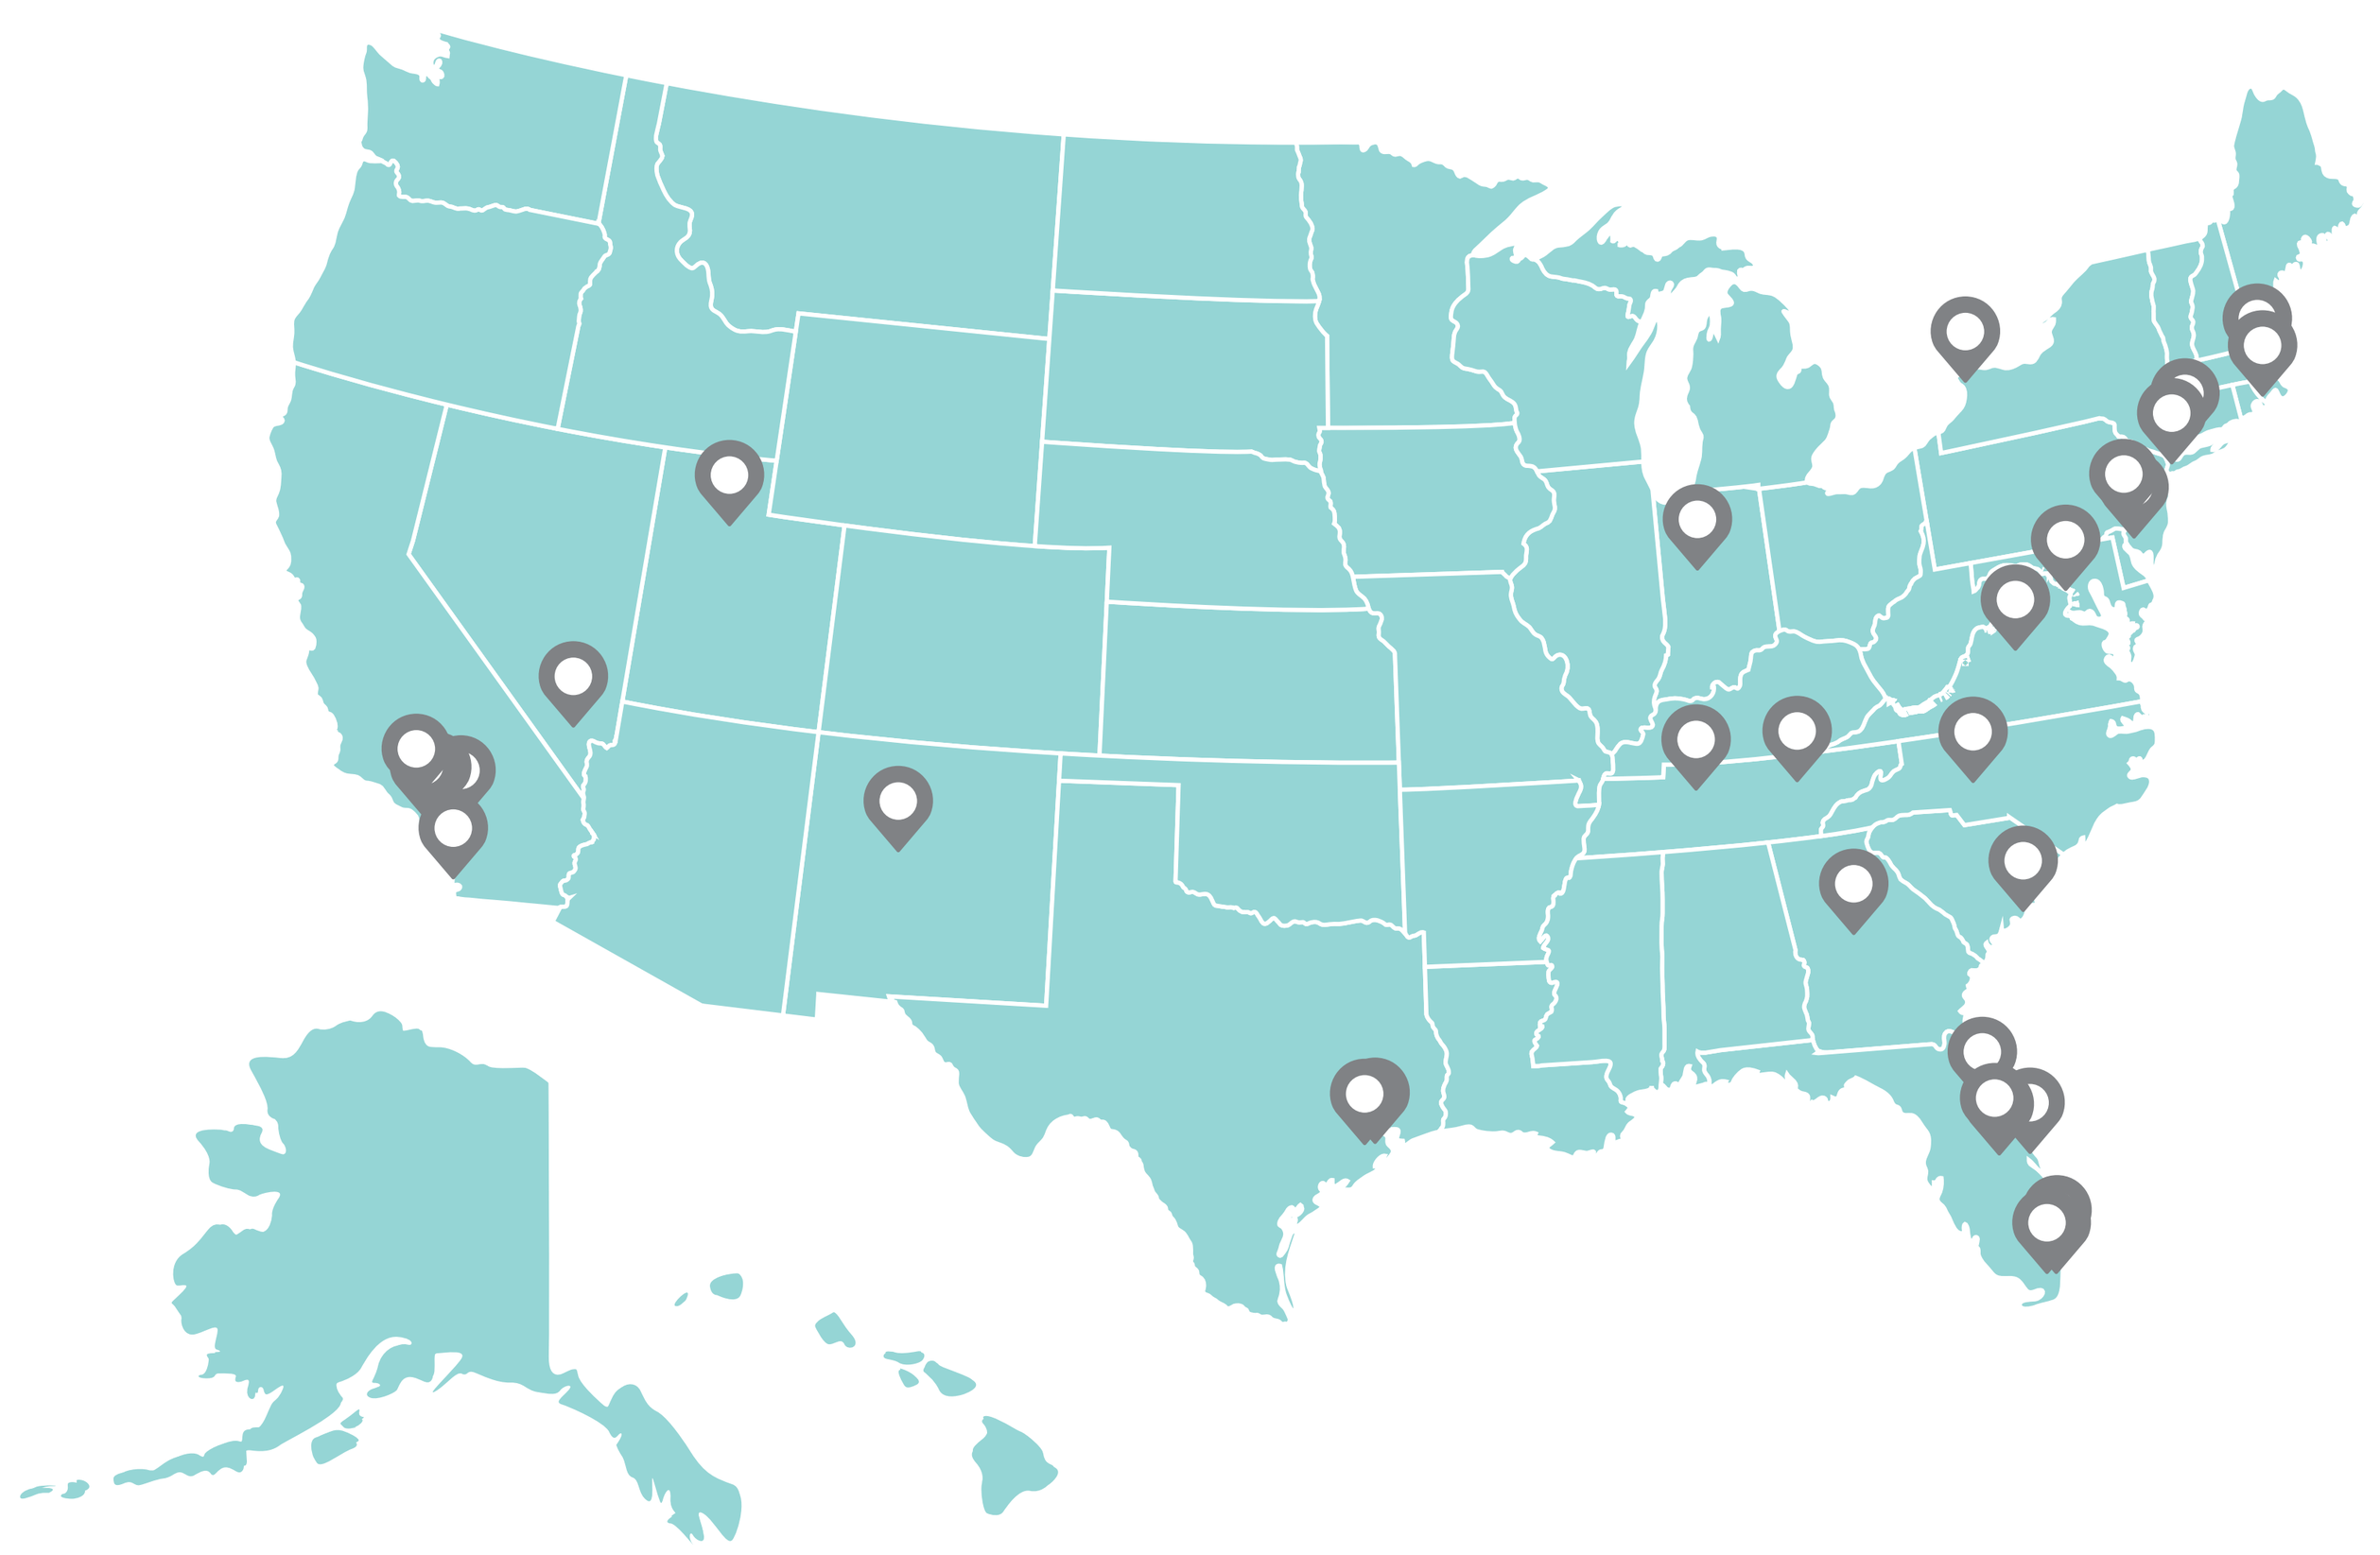

- Study completion is anticipated in December 2024.

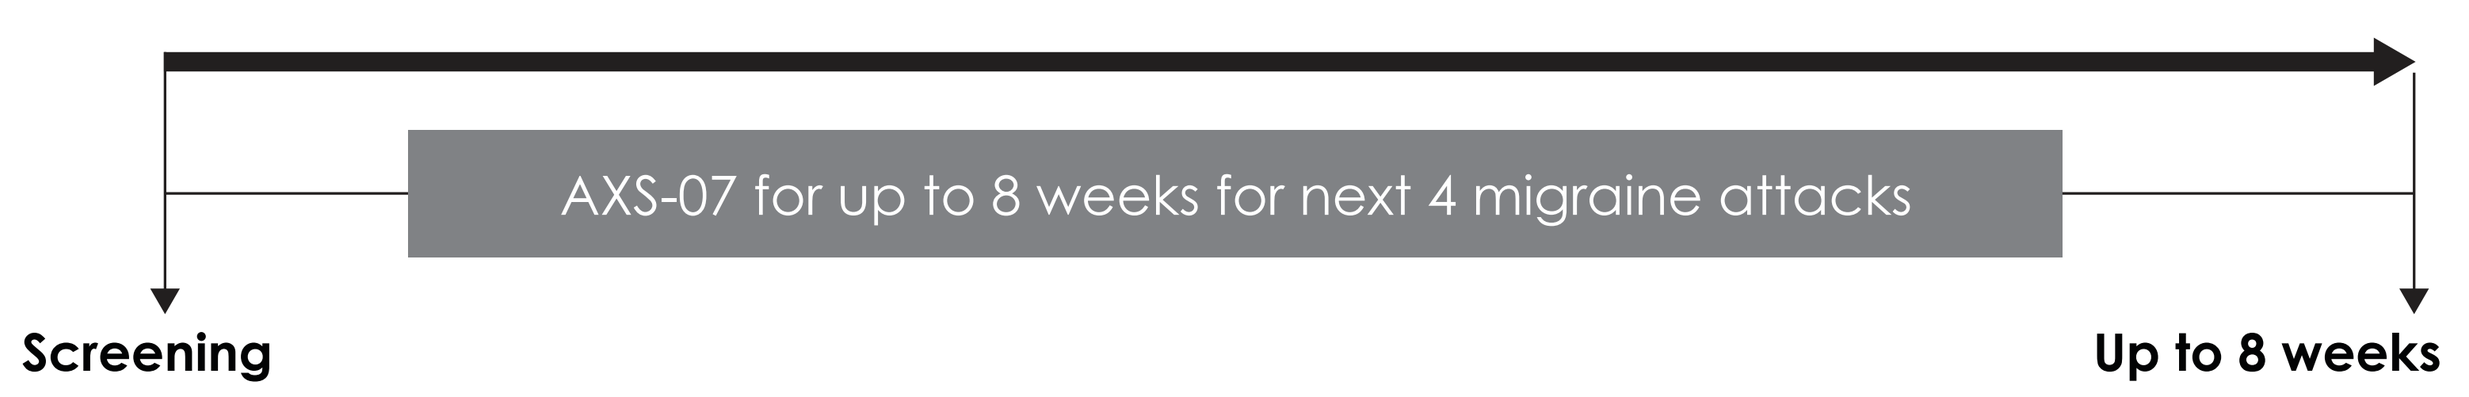
AXS-07 for up to 8 weeks for next 4 migraine attacks
Screening
Up to 8 weeks

Source: https://www.clinicaltrials.gov/study/NCT05550207

Lay Summary

- AXS-07 is a novel medicine that is being developed for acute treatment of migraine pain. It is a single pill taken by mouth containing 2 active ingredients called MoSEIC™ meloxicam and rizatriptan. AXS-07 works by targeting multiple causes of migraine pain.
- Acute treatment with AXS-07 improved migraine pain and associated symptoms compared with an inactive pill ("placebo") and its individual parts (MoSEIC™ meloxicam and rizatriptan) in separate studies involving patients with migraine headaches.
- EMERGE will be the first study to investigate how well AXS-07 works for acute treatment of migraines in patients with a history of migraine attacks and who did not respond well to oral calcitonin gene-related peptide (CGRP) inhibitors.
- EMERGE began enrolling patients in August 2022. Completion of the study is anticipated in December 2024.

References

1. Bigal, et al. *Headache*. 2007;47:475-79.
2. Smelt AFH, et al. *PLoS One*. 2014 Jun 16;9:e98933.
3. Malik SN, et al. *Headache*. 2006;46:773-80.
4. Moreno-Ajona D, et al. *J Clin Med*. 2022;11:1656.
5. AXS-07: A multi-mechanistic approach to treating migraine. Available at: https://www.axsome.com/axs-portfolio/pipeline/about-axs-07. Accessed March 19, 2024.
6. O'Gorman, et al. Poster at 19th Congress of the International Headache Society 2019. IHC-PO-124.
7. Jones, et al. American Academy of Neurology Science Highlights. Emerging Science Presentation, 2020.
8. Jones A, et al. Poster presented at American Academy of Neurology 2022. P13-005.
9. Jones A, et al. Oral presentation at American Headache Society: Virtual Scientific Meeting 2021.